Can Tibial Reference Lines Common to Mechanically Aligned TKA Be Used to Set Tibial Component Rotation in Kinematically Aligned TKA?

Abheetinder S. Brar[1], Joshua D. Roth[1], Stephen M. Howell[1,2], Maury L. Hull[1,2,3]

[1]Biomedical Engineering Graduate Group; [2]Department of Mechanical Engineering; [3]Department of Biomedical Engineering, University of California, Davis, Davis, CA

Download poster using QR code

AAOS 2014, NEW ORLEANS, LOUISIANA | POSTER 119 | www.bme.ucdavis.edu/hull/

INTRODUCTION

Total knee arthroplasty (TKA) is a successful procedure for the majority of patients, however 20-25 percent of patients with a mechanically aligned TKA report dissatisfaction with their knee function[1,2]. Correctly setting internal-external rotation of the tibial component is one factor that affects function[3,4].

Setting component rotation is challenging because there is no consensus on the orientation of the ideal sagittal plane of the knee for establishing reference lines on the tibia, and because finding these reference lines intraoperatively is unreliable[5,6].

In normal knee function, the tibia and patella flex and extend about two transverse axes perpendicular to the sagittal kinematic plane[7,8]. If the goal is to restore normal knee function after TKA, then the ideal plane for setting the rotation of the tibial component is the sagittal kinematic plane[8-10].

Kinematically aligned TKA is an alternative alignment method for which patient-reported satisfaction and function at 2 years is better and revisions at 3 years are fewer than mechanically aligned TKA[11,12]. The goal for setting the rotation of the AP axis of the tibial component in kinematically aligned TKA is to set it parallel to the sagittal kinematic plane[13].

Five tibial reference lines have been used in mechanically-aligned TKA to set rotation of the tibial component[5].

PURPOSE

The present study determined whether any of these five tibial reference lines set the rotation of the tibial component parallel to the sagittal kinematic plane.

METHODS AND MATERIALS

1. Image analysis software was used to create a line parallel to the sagittal kinematic plane on the tibia in fifty three-dimensional (3-D) bone models of normal lower extremities from white subjects using a four-step alignment algorithm (Figure 1).

2. Eight landmarks were identified on each tibia (Figure 2).

3. Five tibial reference lines were drawn by connecting two landmarks and were termed (shown later in Figure 3):
 1. Reference line connecting medial border of the tibial tubercle with the center of the PCL fossa.
 2. Reference line connecting medial 1/3rd of the tibial tubercle with center of the PCL fossa.
 3. Reference line connecting most anterior point of the tibial tubercle with center of the PCL fossa.
 4. Reference line perpendicular to the line connecting the center of each tibial condyle.
 5. Reference line perpendicular to the line connecting the most posterior point on each tibial condyle.

4. The angle that each tibial reference line formed with the line parallel to the sagittal kinematic plane quantified the component rotation. A positive value indicated external rotation of that tibial reference line from the sagittal kinematic plane.

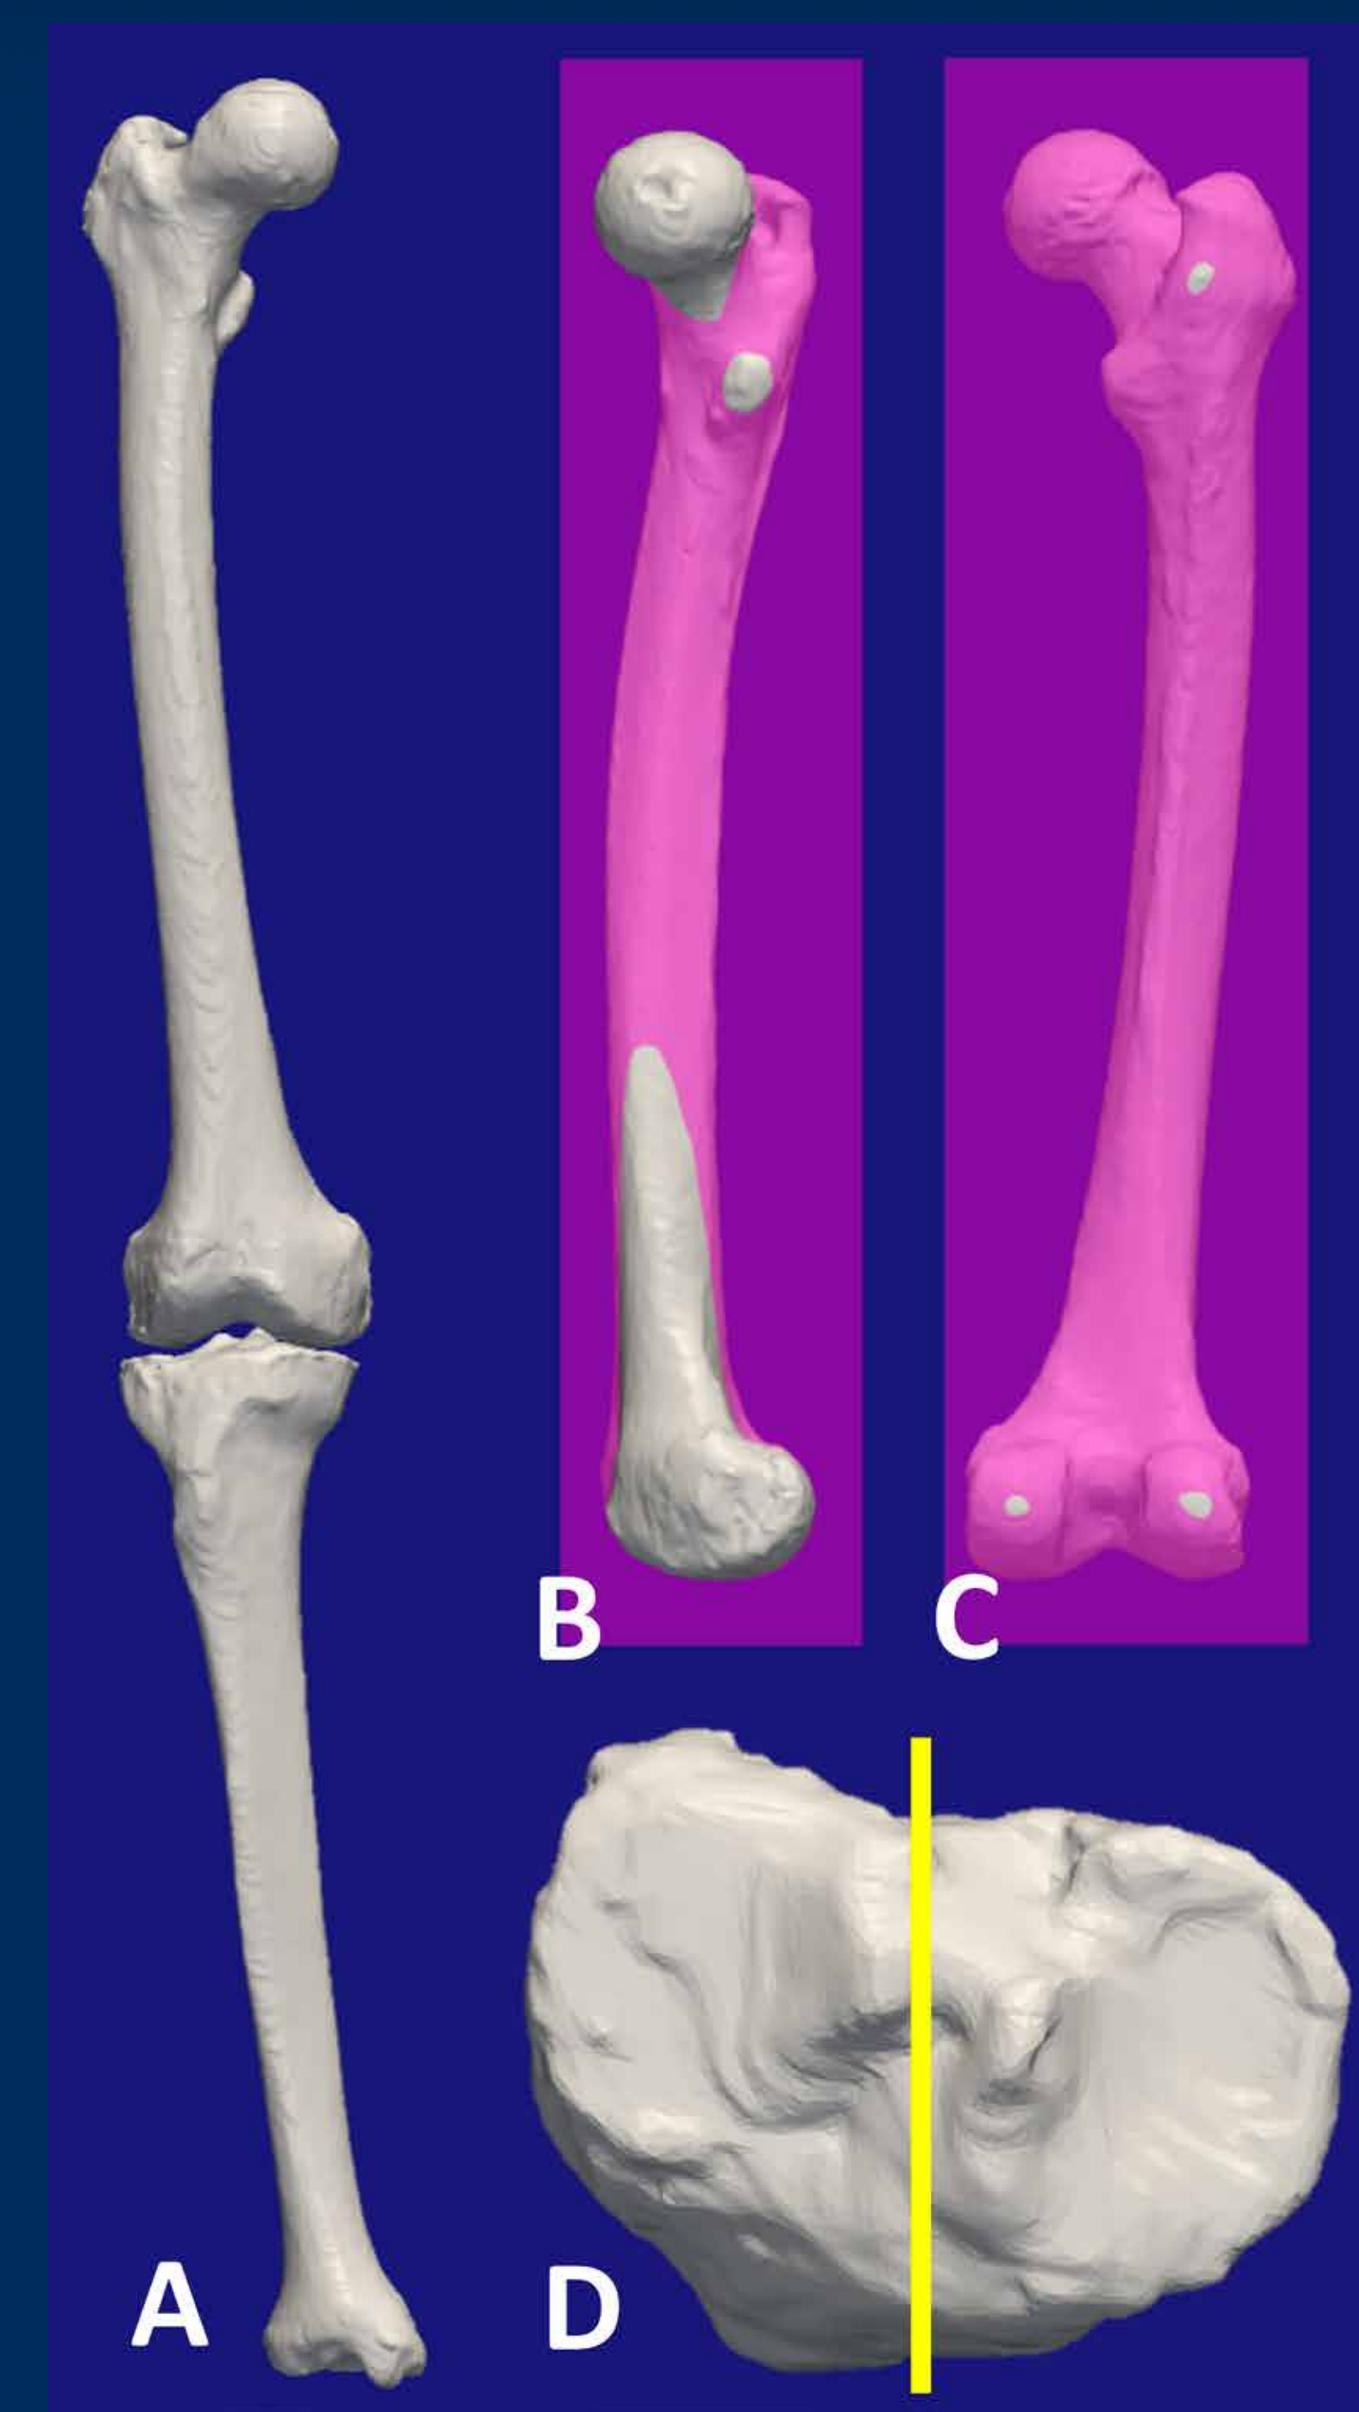
A
B
C
D

Figure 1. The composite shows a 3-D model of a right lower extremity and the four steps for orienting the extremity in the three kinematic planes.

A. The bone model was imported into software.

B. The tibia was hidden, and the femur was projected in the sagittal kinematic plane by superimposing the medial and lateral femoral condyles.

C. The femur was projected in the coronal kinematic plane and perpendicular to the sagittal kinematic plane by placing the most posterior point of each femoral condyle and greater trochanter tangent to a surface.

D. The transformations applied to the femur were applied to the tibia, the tibia was projected perpendicular to the other two planes in the axial kinematic plane, and the line parallel to the sagittal kinematic plane (yellow) was drawn on the proximal articular surface of the tibia.

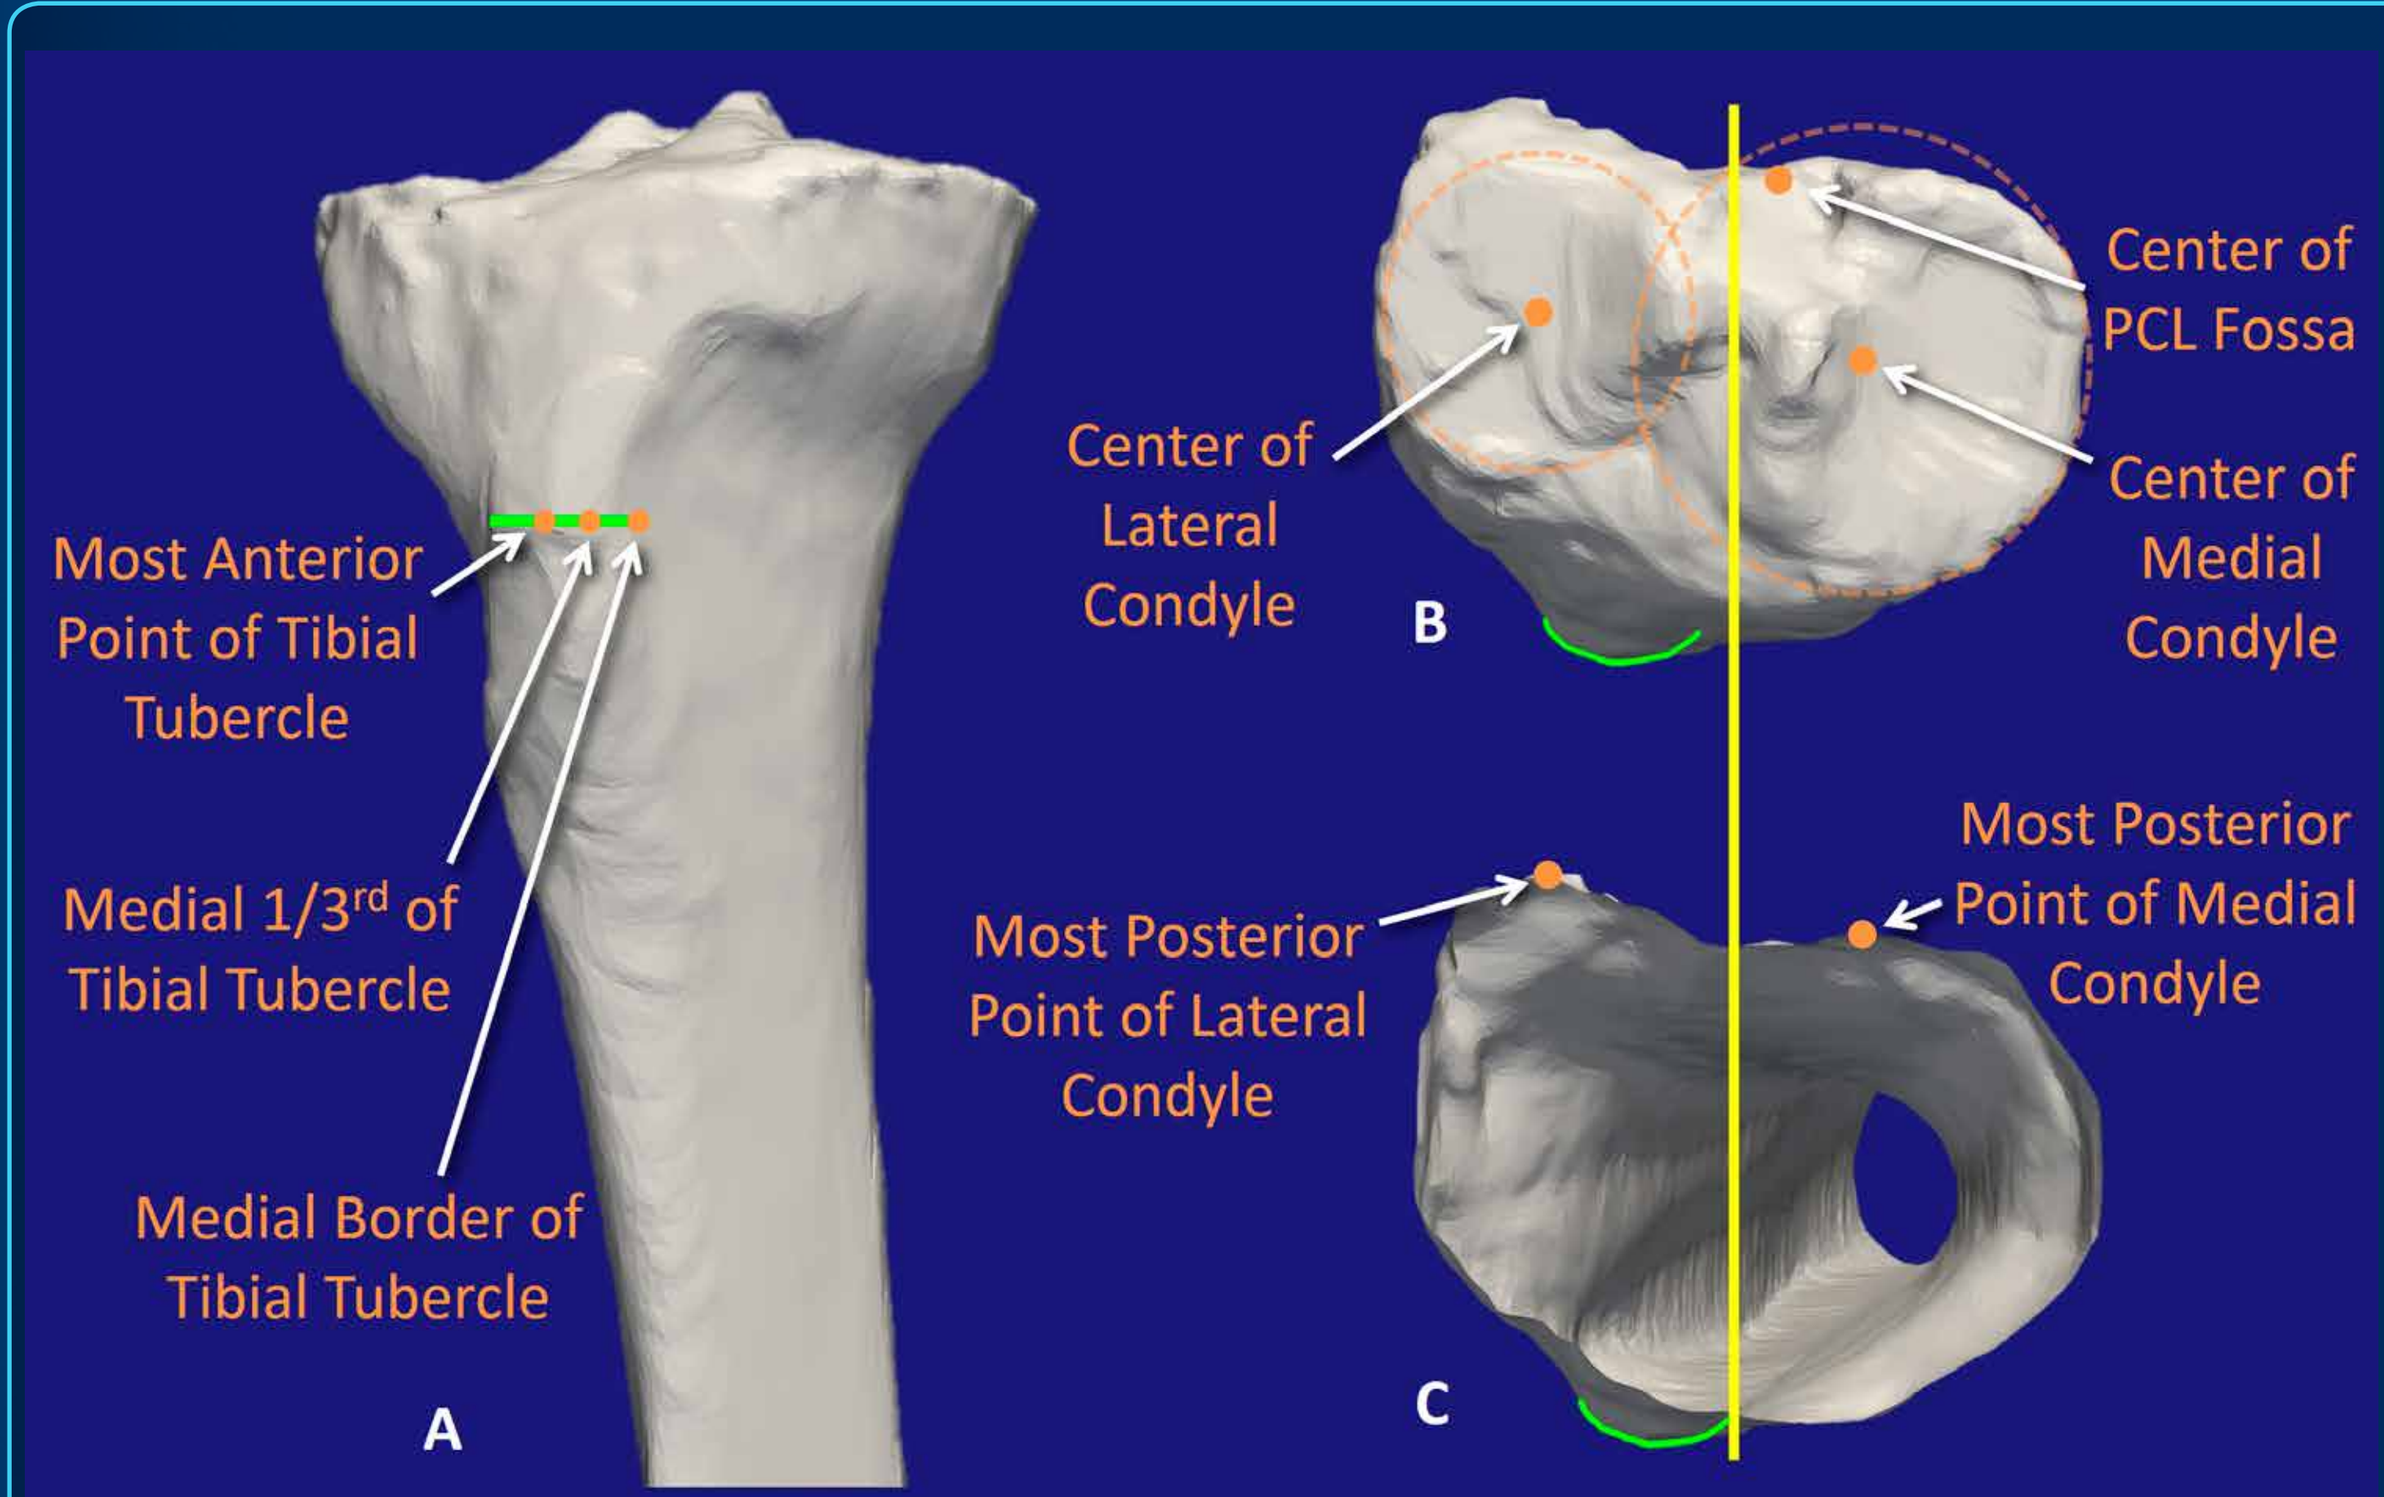
Most Anterior Point of Tibial Tubercle
Medial 1/3rd of Tibial Tubercle
Medial Border of Tibial Tubercle
A
Center of Lateral Condyle
B
Center of PCL Fossa
Center of Medial Condyle
Most Posterior Point of Lateral Condyle
Most Posterior Point of Medial Condyle
C

Figure 2. A composite of a right tibia shows the eight landmarks for constructing five tibial reference lines used in mechanically aligned TKA.

A. The most anterior point, medial border, and medial 1/3rd of the tibial tubercle (green arc), were identified on the projection of the tibia in the coronal kinematic plane.

B. The center of the PCL fossa and the center of the medial and lateral tibial condyles were identified on the axial kinematic plane of the proximal articular surface of the tibia.

C. The most posterior points on the medial and lateral condyles were identified 10 mm distal to the deepest portion of the medial tibial condyle, which shows the hollow cavity of the cortical bone. The yellow line is parallel to the sagittal kinematic plane.

RESULTS

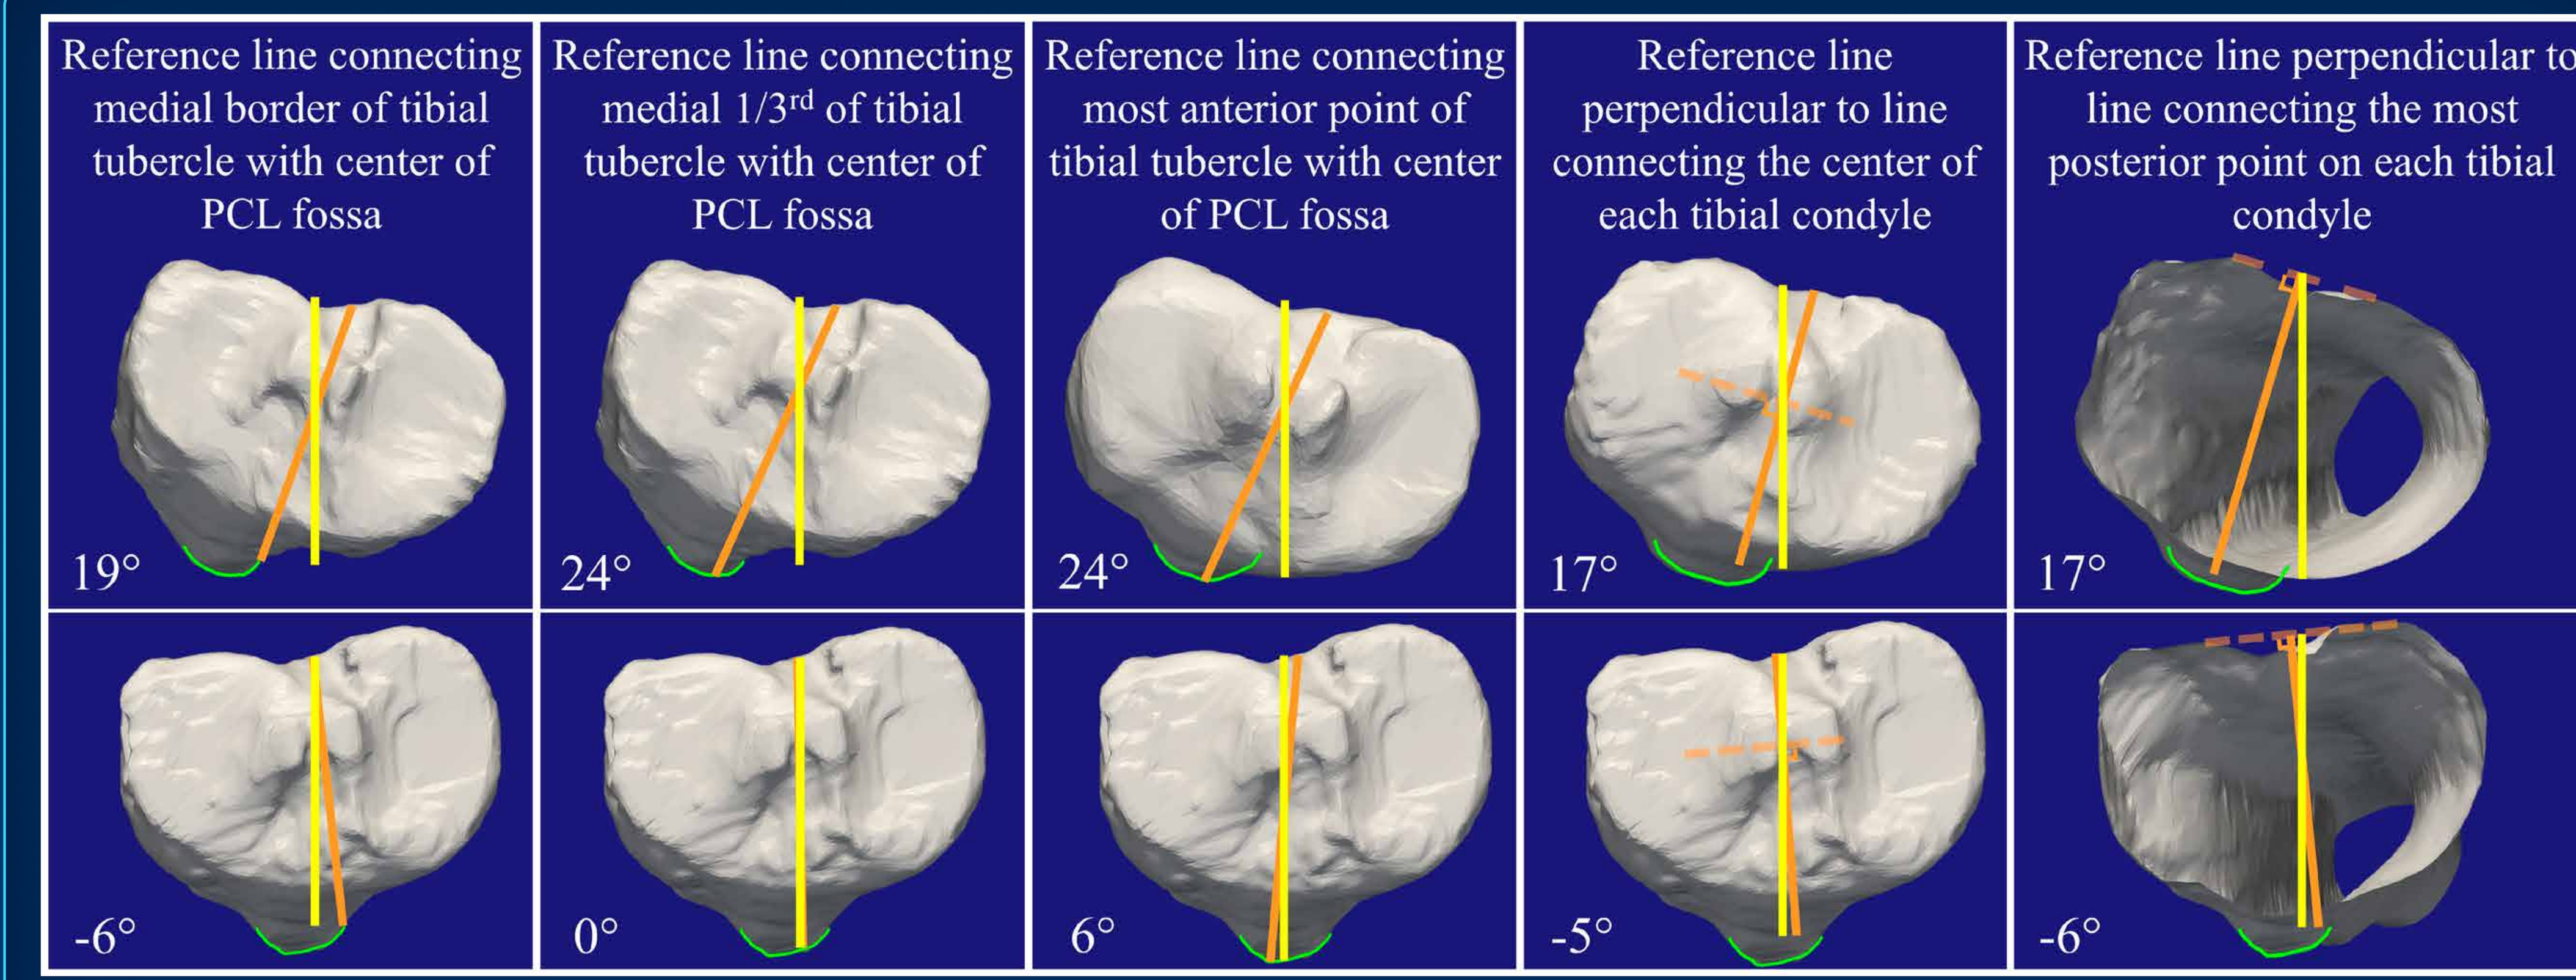
Reference line connecting medial border of tibial tubercle with center of PCL fossa
Reference line connecting medial 1/3rd of tibial tubercle with center of PCL fossa
Reference line connecting most anterior point of tibial tubercle with center of PCL fossa
Reference line perpendicular to line connecting the center of each tibial condyle
Reference line perpendicular to line connecting the most posterior point on each tibial condyle
19°
24°
24°
17°
17°
-6°
0°
6°
-5°
-6°

Figure 3. The composite shows the maximum external (positive) and internal (negative) rotation of each tibial reference line (orange) from the sagittal kinematic plane (yellow). Each tibia is viewed as right. The green arc outlines the tibial tubercle. The smallest range of rotation was 22°.

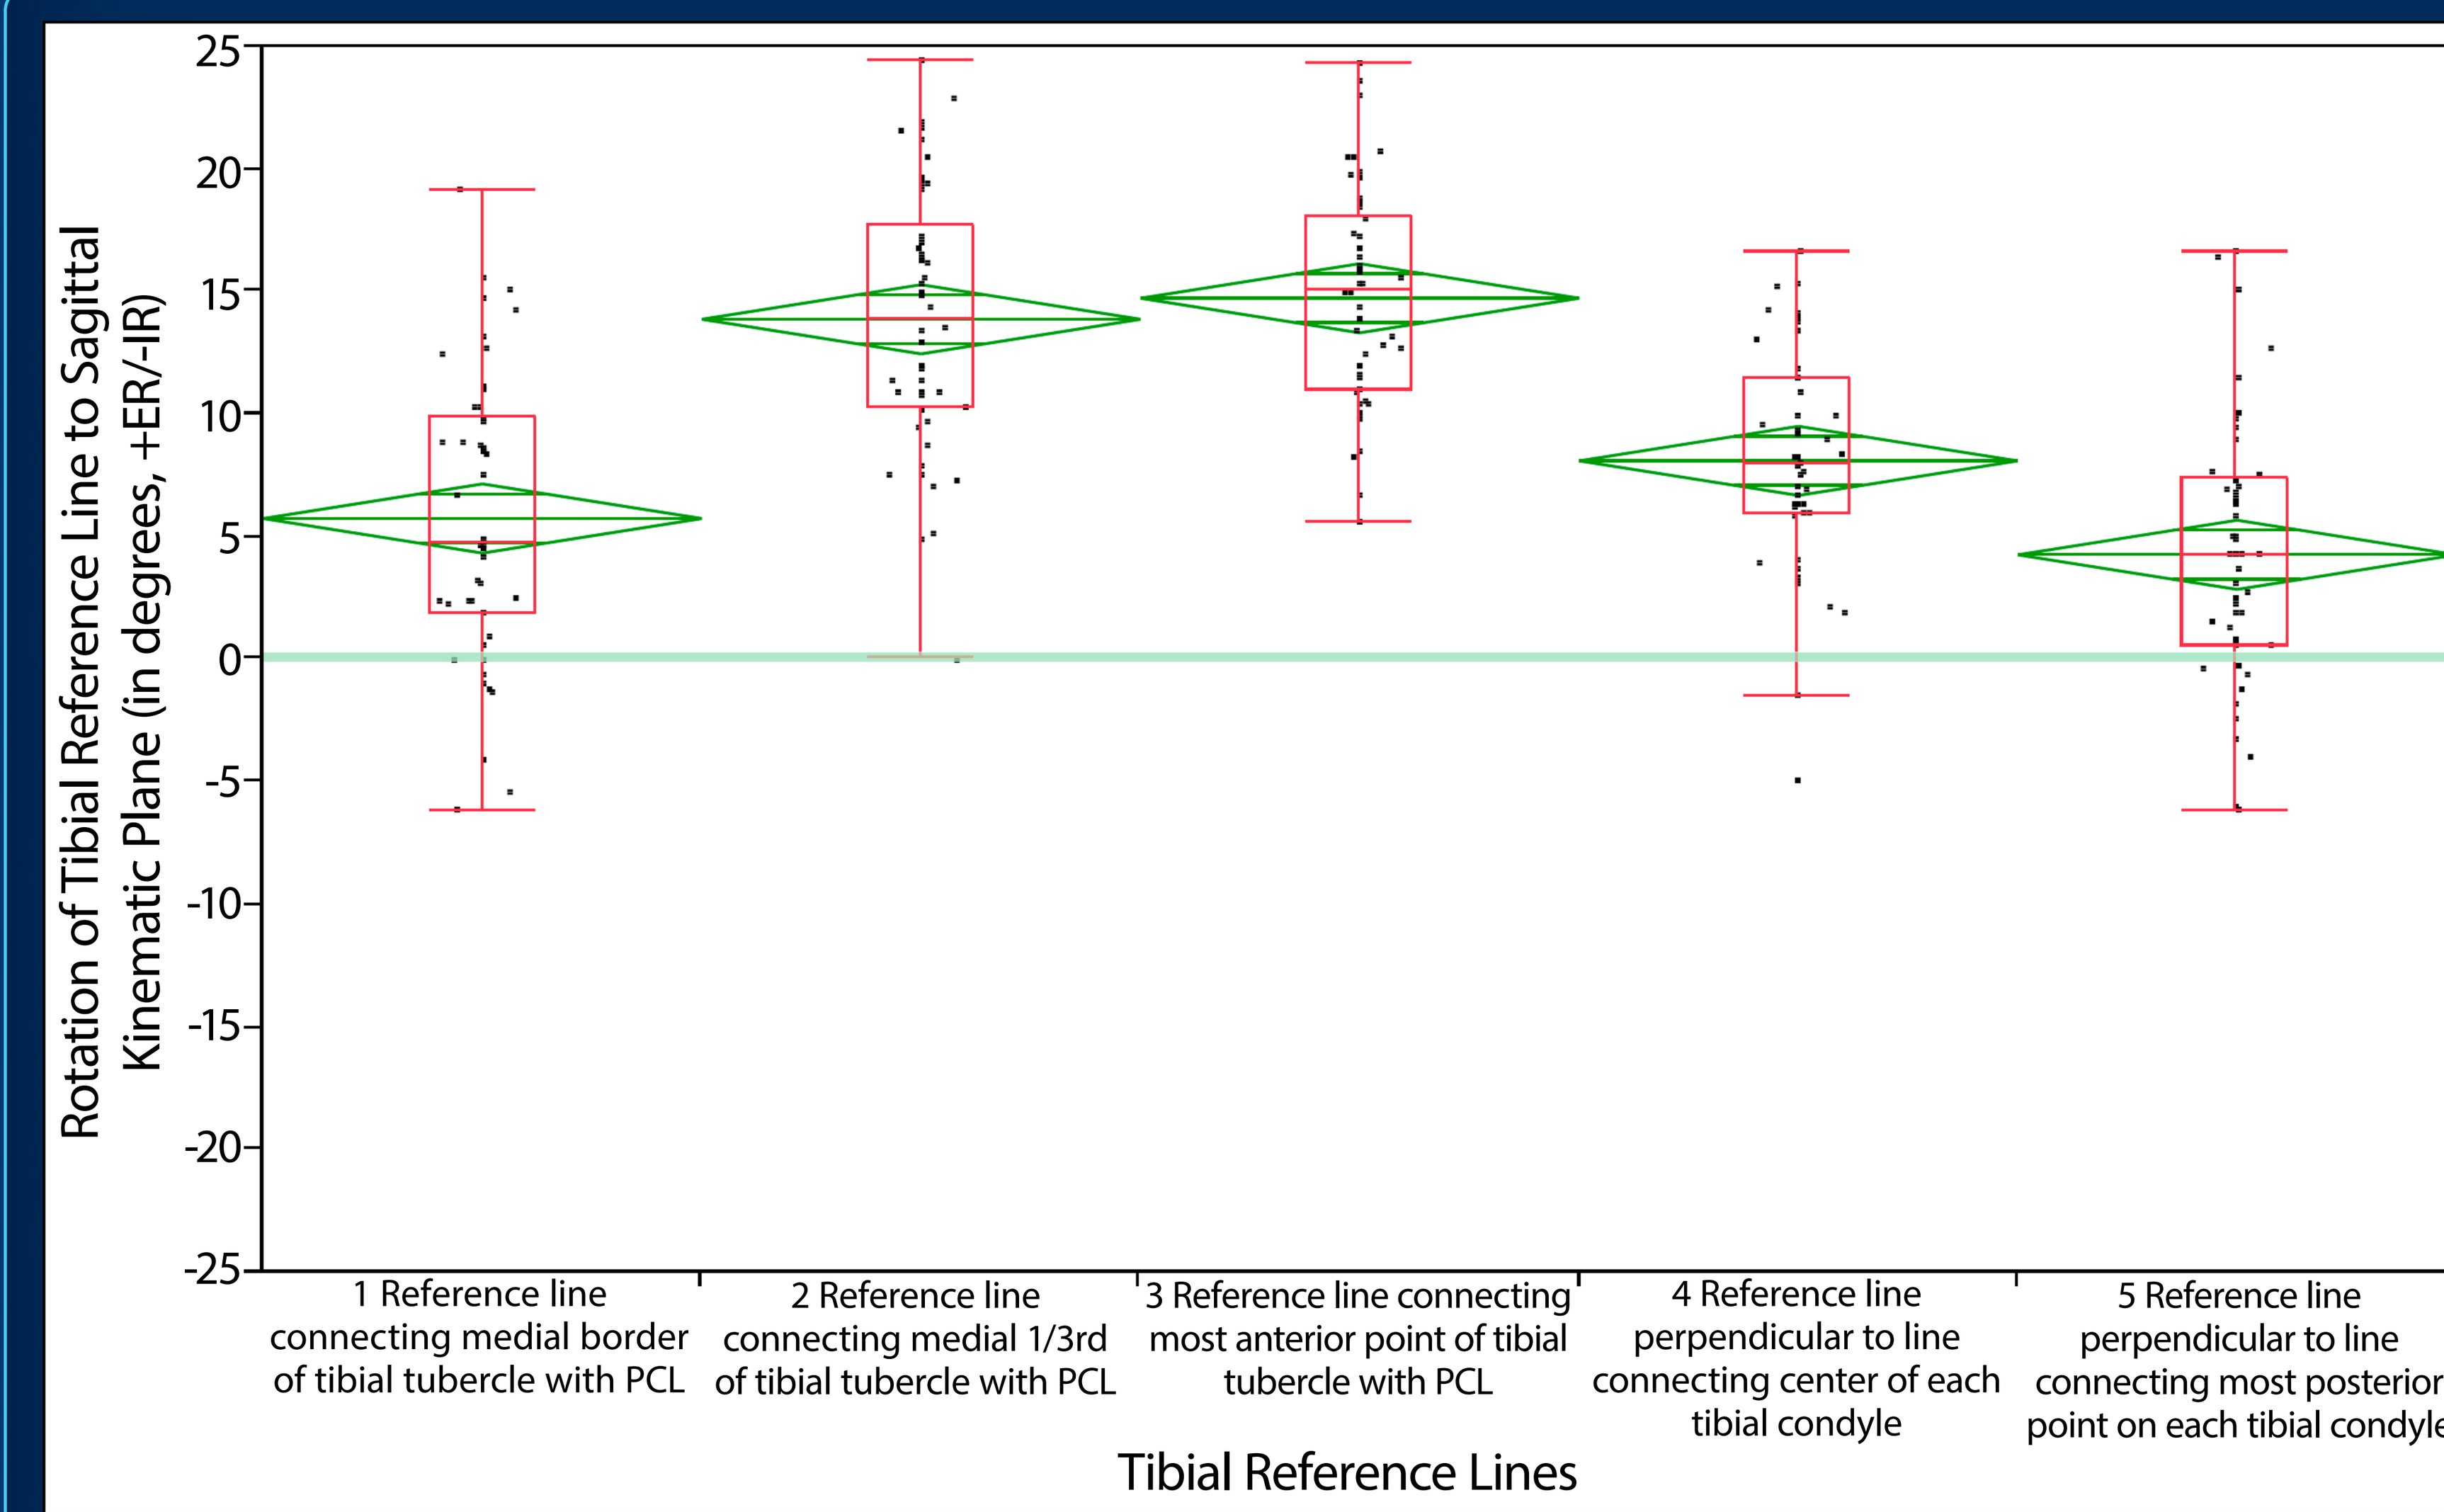
Rotation of Tibial Reference Line to Sagittal Kinematic Plane (in degrees, +ER/-IR)
25
20
15
10
5
0
-5
-10
-15
-20
-25
1 Reference line connecting medial border of tibial tubercle with PCL
2 Reference line connecting medial 1/3rd of tibial tubercle with PCL
3 Reference line connecting most anterior point of tibial tubercle with PCL
4 Reference line perpendicular to line connecting center of each tibial condyle
5 Reference line perpendicular to line connecting most posterior point on each tibial condyle
Tibial Reference Lines

Figure 4. The graph shows the mean and the upper and lower 95% confidence limits (green diamond) of the rotation of each tibial reference line from the sagittal kinematic plane. The average rotation of each tibial reference line ranged from 4° to 15° external to the sagittal kinematic plane. The 95% confidence interval for all 5 tibial reference lines did not include 0°. On average none of the five tibial reference lines were parallel to the sagittal kinematic plane.

DISCUSSION

Our study shows that the five tibial reference lines common to mechanically aligned TKA externally rotate the tibial component from the sagittal kinematic plane. The surgeon can expect a wide range of component rotation when using a reference line that references the tibial tubercle because there is wide variability in the medial-lateral location of the tibial tubercle with respect to the medial border of the tibia[14]. Accordingly, new methods that accurately set rotation of the tibial component in kinematically aligned TKA should be developed.

1. Baker PN, van der Meulen JH, Lewsey J, Gregg PJ, National Joint Registry for E, Wales: The role of pain and function in determining patient satisfaction after total knee replacement. Data from the National Joint Registry for England and Wales. J Bone Joint Surg Br 2007;89:893-900.
2. Bourne RB, Chesworth BM, Davis AM, Mahomed NN, Charron KDJ: Patient satisfaction after total knee arthroplasty: who is satisfied and who is not? Clinical orthopaedics and related research 2010;468:57-63.
3. Berger RA, Crossett LS, Jacobs JJ, Rubash HE: Malrotation causing patellofemoral complications after total knee arthroplasty. Clinical orthopaedics and related research 1998;356:144-153.
4. Lutzner J, Kirschner S, Gunther KP, Harman MK: Patients with no functional improvement after total knee arthroplasty show different kinematics. International orthopaedics 2012;36:1841-1847.
5. Siston RA, Goodman SB, Patel JJ, Delp SL, Giori NJ: The high variability of tibial rotational alignment in total knee arthroplasty. Clinical orthopaedics and related research 2006;452:65-69.
6. Cobb JP, Dixon H, Dandachli W, Iranpour F: The anatomical tibial axis: reliable rotational orientation in knee replacement. J Bone Joint Surg Br 2008;90:1032-1038.
7. Coughlin KM, Incavo SJ, Churchill DL, Beynnon BD: Tibial axis and patellar position relative to the femoral epicondylar axis during squatting. The Journal of arthroplasty 2003;18:1048-1055.
8. Hollister AM, Jatana S, Singh AK, Sullivan WW, Lupichuk AG: The axes of rotation of the knee. Clinical orthopaedics and related research 1993;290:259-268.
9. Eckhoff D, Hogan C, DiMatteo L, Robinson M, Bach J: Difference between the epicondylar and cylindrical axis of the knee. Clinical orthopaedics and related research 2007;461:238-244.
10. Gu Y, Roth J, Howell SM, Hull ML: How frequently do four methods for mechanically-aligning a total knee arthroplasty cause collateral ligament imbalance and change alignment from normal in Caucasians? The Journal of bone and joint surgery In Press.
11. Dossett H, Swartz G, Estrada N, Lefevre G, Kwasman B: Kinematically versus mechanically aligned total knee arthroplasty. Orthopedics 2012;35:160-169.
12. Howell SM, Howell SJ, Kuznik KT, Cohen J, Hull ML: Does a kinematically aligned total knee arthroplasty restore function without failure regardless of alignment category? Clinical orthopaedics and related research 2012.
13. Nedopil A, Howell S, Rudert M, Roth J, Hull M: How Frequent is Rotational Mismatch Within 0 ± 10 degrees in Kinematically-Aligned TKA? . Orthopedics 2013;36:e1515-e1520.
14. Howell SM, Chen J, Hull ML: Variability of the location of the tibial tubercle affects the rotational alignment of the tibial component in kinematically aligned total knee arthroplasty. Knee surgery, sports traumatology, arthroscopy : official journal of the ESSKA 2013;21:2288-2295.